Poster# 1669

Tufts Medical Center

IDWeek 2022

A US-Based National Surveillance Study for the Susceptibility and Epidemiology of *Clostridioides difficile* Associated Diarrheal Isolates with Special Reference to Ridinilazole: 2020-2021

Summit therapeutics

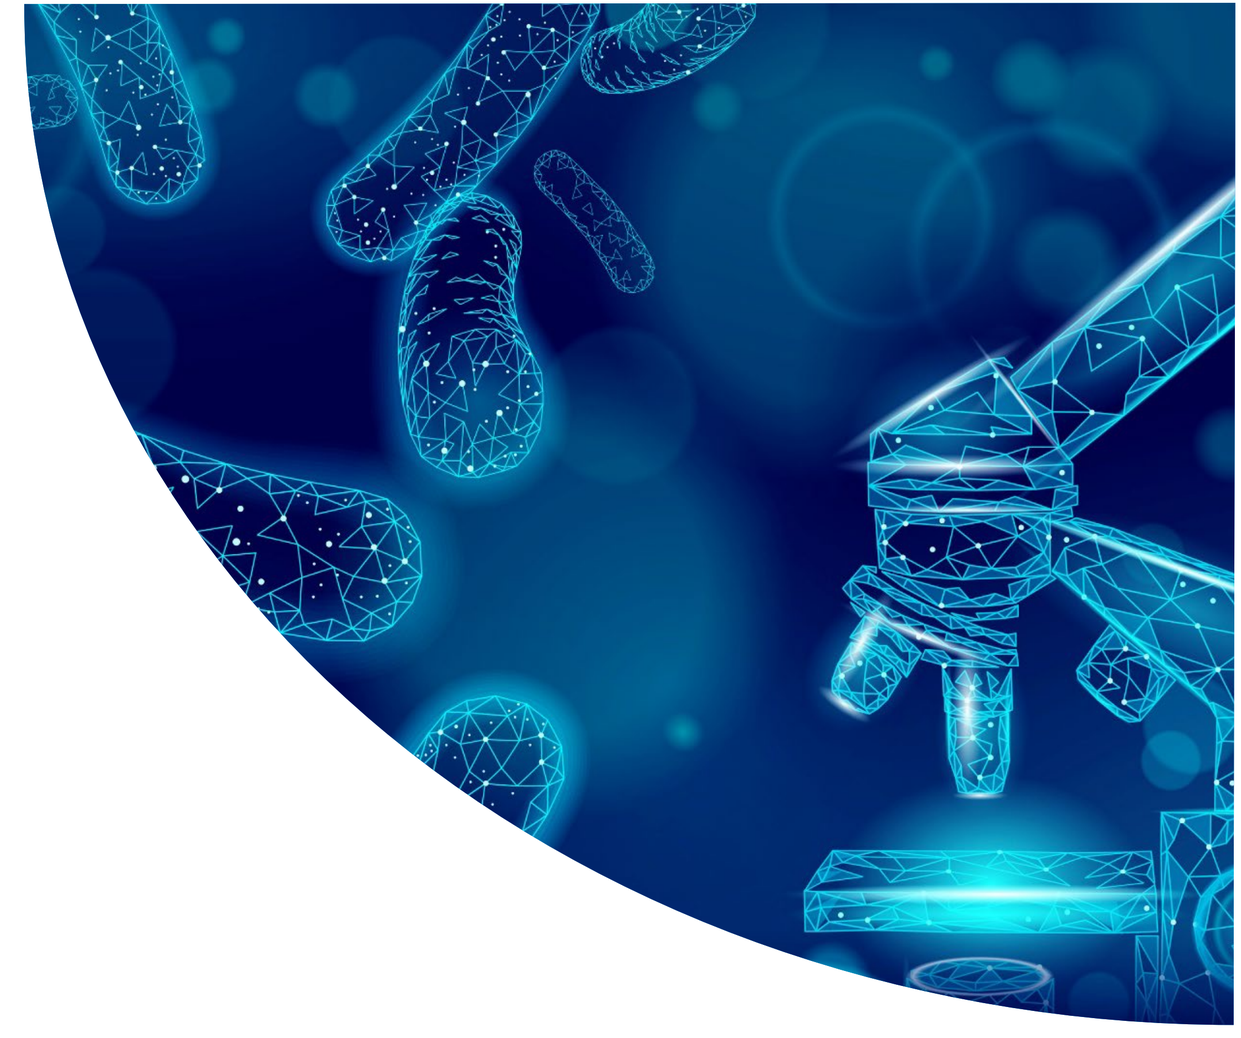

D.R. Snydman[1,2], L.A. McDermott[1,2], C.M. Thorpe[1,2], E.J.C. Goldstein[3], A. N. Schuetz[4], S. Johnson[5], D.N. Gerding[5], L. Gluck[6] , D. Bourdas[6] , K. C. Carroll[7] , C.K. Lancaster [8], K.W. Garey[8], S. T. Walk[9] , E. Duperchy[10]

[1]Tufts Medical Center, [2]Tufts School of Medicine, Boston, MA; [3]RKindred Health Care System, Rancho Cucamonga, CA; [4]Mayo Clinic School of Medicine and Science, Rochester, MN; [5]Hines VA Hospital, Hines, IL; [6]Johns Hopkins Hospital, Baltimore, MD; [7]Johns University School of Medicine, Baltimore, MD; [8]University of Houston College of Pharmacy, Houston, TX; [9]Montana State University, Bozeman, MT; [10]Summit Therapeutics, Abingdon, UK

Presenting Author: D.R Snydman, Email: dsnydman@tuftsmedicalcenter.org , Tel: +1 617-636-5788

BACKGROUND

- *Clostridioides* (formerly Clostridium) *difficile* infection (CDI) continues to be a problem worldwide causing substantial morbidity and mortality. It is currently among the top five threats in antimicrobial resistance in the United States listed by the Centers for Disease Control (CDC).[1]
- The appearance of fluoroquinolone-resistant NAP1/BI/027 isolates, associated with epidemics of complicated CDI cases, toxic megacolon and increased mortality, has only further highlighted the urgency of our need to understand the epidemiology of *C. difficile* in the US.[7]
- We have previously been involved with national surveillance of *C. difficile* susceptibility to a panel of agents from 2011-2016.[2,3]
- Ridinilazole is in development for treatment of *C. difficile* associated diarrhea with a potentially narrower spectrum than fidaxomicin.[4,5,6]
- We collected 300 isolates of *C. difficile* from 6 different medical centers in the United States which are geographically separated to conduct in vitro antimicrobial susceptibility testing against ridinilazole and comparators.
- Ribotyping was also performed on all isolates.

MATERIALS

- A convenience sample of either stools or isolates from patients diagnosed with C. *difficile* infection were referred to the Special Studies Laboratory at Tufts Medical Center across 6 medical centers.
- Thawed stool samples were ethanol shocked prior to being plated. Culture of stool and confirmation of the isolates as C. *difficile* was accomplished by plating on C. *difficile* selective medium (cycloserine-cefoxitin-fructose agar with taurocholate, Anaerobe Systems) and observing for characteristic colonial morphology. A proline disc test (Remel Products) and gram stain were performed. This was followed by using API20A® (BioMerieux Inc).
- Susceptibility Testing against the panel of antibiotics shown in Table 1 was performed in singlicate using the Agar Dilution Method from CLSI, M11-A9.
- The rates of resistance of the antimicrobial agents were determined using currently accepted CLSI breakpoints for anaerobes. For agents that do not have CLSI recommendations, or FDA recommendations, the manufacturer's proposed breakpoint(s) were used (Table 1). We also looked at rates of resistance using EUCAST breakpoints, based on epidemiologic cut-off values, which have been established for C. *difficile*.
- Ribotyping of all isolates was performed at the Walk Lab.
- Results were entered into the Montana State University Walk Lab pipeline for analysis and matching to known ribotypes by generating (Bray-Curtis similarity indices http://walklab.rcg.montana.edu).

Table 1: Antimicrobial Agents Tested, Ranges and Breakpoints for Susceptibility

Antimicrobial Agent	Abbreviation	Range Tested (µg/ml)	Breakpoint (µg/ml)	
			CLSI	EUCAST
Ridinilazole	RDZ	4 - 0.004	NA[1]	NA[1]
Vancomycin	VAN	32 - 0.25	≥4[2]	>2[2]
Fidaxomicin	FDX	4 - 0.004	NA[1]	NA[1]
Metronidazole	MTZ	16 - 0.06	≥32	>2[2]
Clindamycin	CLI	32 - 0.5	≥8	NA
Imipenem	IMI	16 - 0.12	≥16	NA[1]
Moxifloxacin	MOX	32 - 0.5	≥8	>4[2]
Rifampin	RIF	4 - 0.004	NA[1]	>0.004[2]
Rifaximin	RFX	4 - 0.004	NA[1]	NA[1]
Tigecycline	TGC	4 - 0.004	≥16[3]	>0.25[2]

[1]NA:not applicable, CLSI or EUCAST recommended breakpoint for resistance not available.
[2]The CLSI or EUCAST, as applicable, epidemiologic cut-off value was applied, in the absence of a clinical breakpoint.
[3]For tigecycline the breakpoint for resistance recommended for anaerobes by the FDA was used.

Ridinazole is an investigational compound that is not approved by any regulatory body.
Learn more at www.summittxinc.com

RESULTS

Ridinilazole had excellent activity against all isolates collected in the US in 2020-2021 with a MIC_{90} of 0.25 µg/ml, MIC_{90} lower than that of the CDI antibiotics vancomycin, fidaxomicin and metronidazole, in this study.

Table 2: MIC (µg/ml) of Ridinilazole and Comparator Antibiotics Against 300 *C. difficile* Isolates

Antimicrobial Agent	MIC Range (µg/ml)	MIC_{50} (µg/ml)	MIC_{90} (µg/ml)	Percent Resistant	
				CLSI	EUCAST
Ridinilazole	**0.03 - 0.5**	**0.25**	**0.25**	**NA[1]**	**NA[1]**
Vancomycin	≤0.25 – 4	2	2	0.7%[2]	0.7%[2]
Fidaxomicin	0.03 - 0.5	0.25	0.5	NA[1]	NA[1]
Metronidazole	0.12 – 4	0.5	1	0.0%	0.3%[2]
Clindamycin	≤0.5 - >32	4	>32	26.0%	NA[1]
Imipenem	2 – 16	4	8	5.0%	NA[1]
Moxifloxacin	1 – 32	2	16	14.7%	14.7%[2]
Rifampin	≤0.004 - >4	≤0.004	0.008	NA[1]	5.9%[2]
Rifaximin	≤0.004 - >4	0.015	0.03	NA[1]	NA[1]
Tigecycline	≤0.06 - 0.5	0.12	0.12	0.0%[3]	0.7%[2]

[1]NA:not applicable, CLSI or EUCAST recommended breakpoint for resistance not available.
[2]The CLSI or EUCAST, as applicable, epidemiologic cut-off value was applied, in the absence of a clinical breakpoint.
[3]For tigecycline the breakpoint for resistance recommended for anaerobes by the FDA was used.

The most common ribotype was 014-020 (14.3% compared to 11.8% 2016), followed by 106 (10%, 15% in 2016), 027 (10%, 13.1% in 2016), 002 (8%, 8.5% in 2016), 078-126 (4.3%, 1.3% in 2016).
Ribotypes 027 and RT 078-126 were the only two hypervirulent ribotypes detected in this study (among 023, 027, 078-126, 176, 198 and 244 hypervirulent ribotypes).

Figure 1. Distribution of Ribotypes for the 300 Isolates

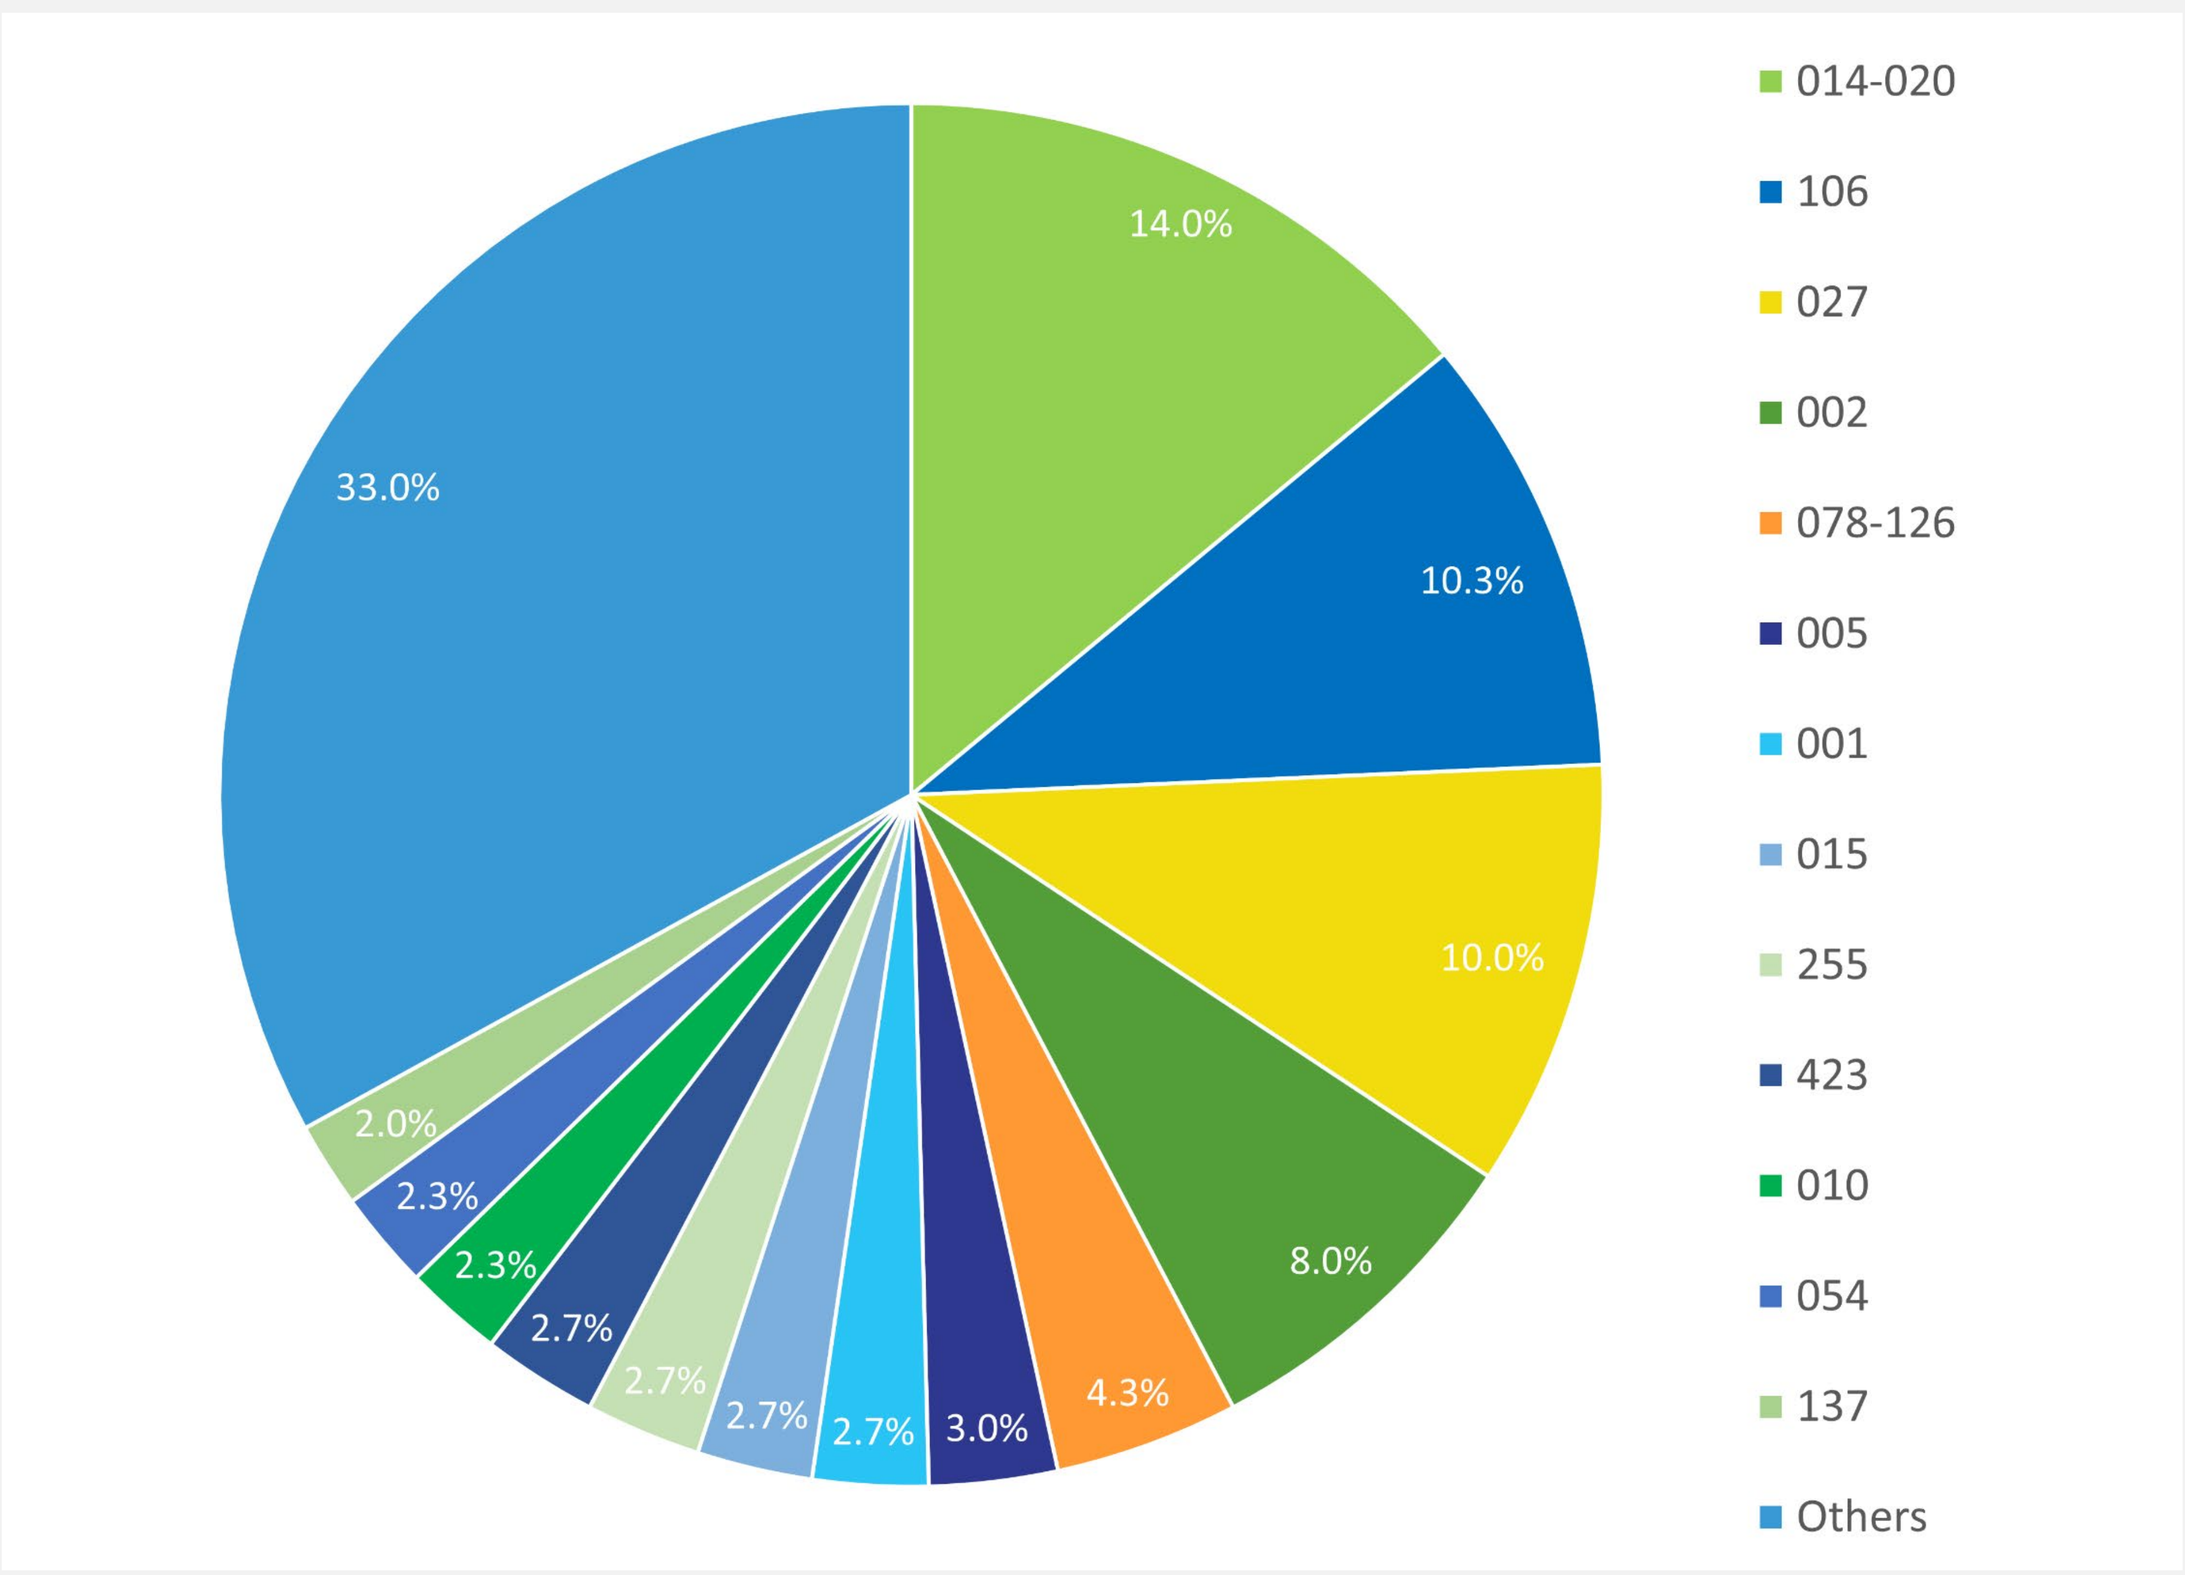

14.0%
10.3%
10.0%
8.0%
4.3%
3.0%
2.7%
2.7%
2.7%
2.7%
2.3%
2.3%
2.0%
33.0%
014-020
106
027
002
078-126
005
001
015
255
423
010
054
137
Others

"Others" includes ribotypes representing < 2% of all *C. difficile* isolates.

Ridinilazole showed a similar MIC range and distribution against the non-hypervirulent ribotype 014-20 and hypervirulent ribotypes 027 and 078-126.
Clindamycin and moxifloxacin showed very different MIC range and distribution.

Figure 2. Frequency of MIC (µg/ml) Distribution of Ridinilazole and Comparator Antibiotics Against All, Non-Hypervirulent (RT 014-20) and Hypervirulent Ribotypes (RT 027 and 078-126)

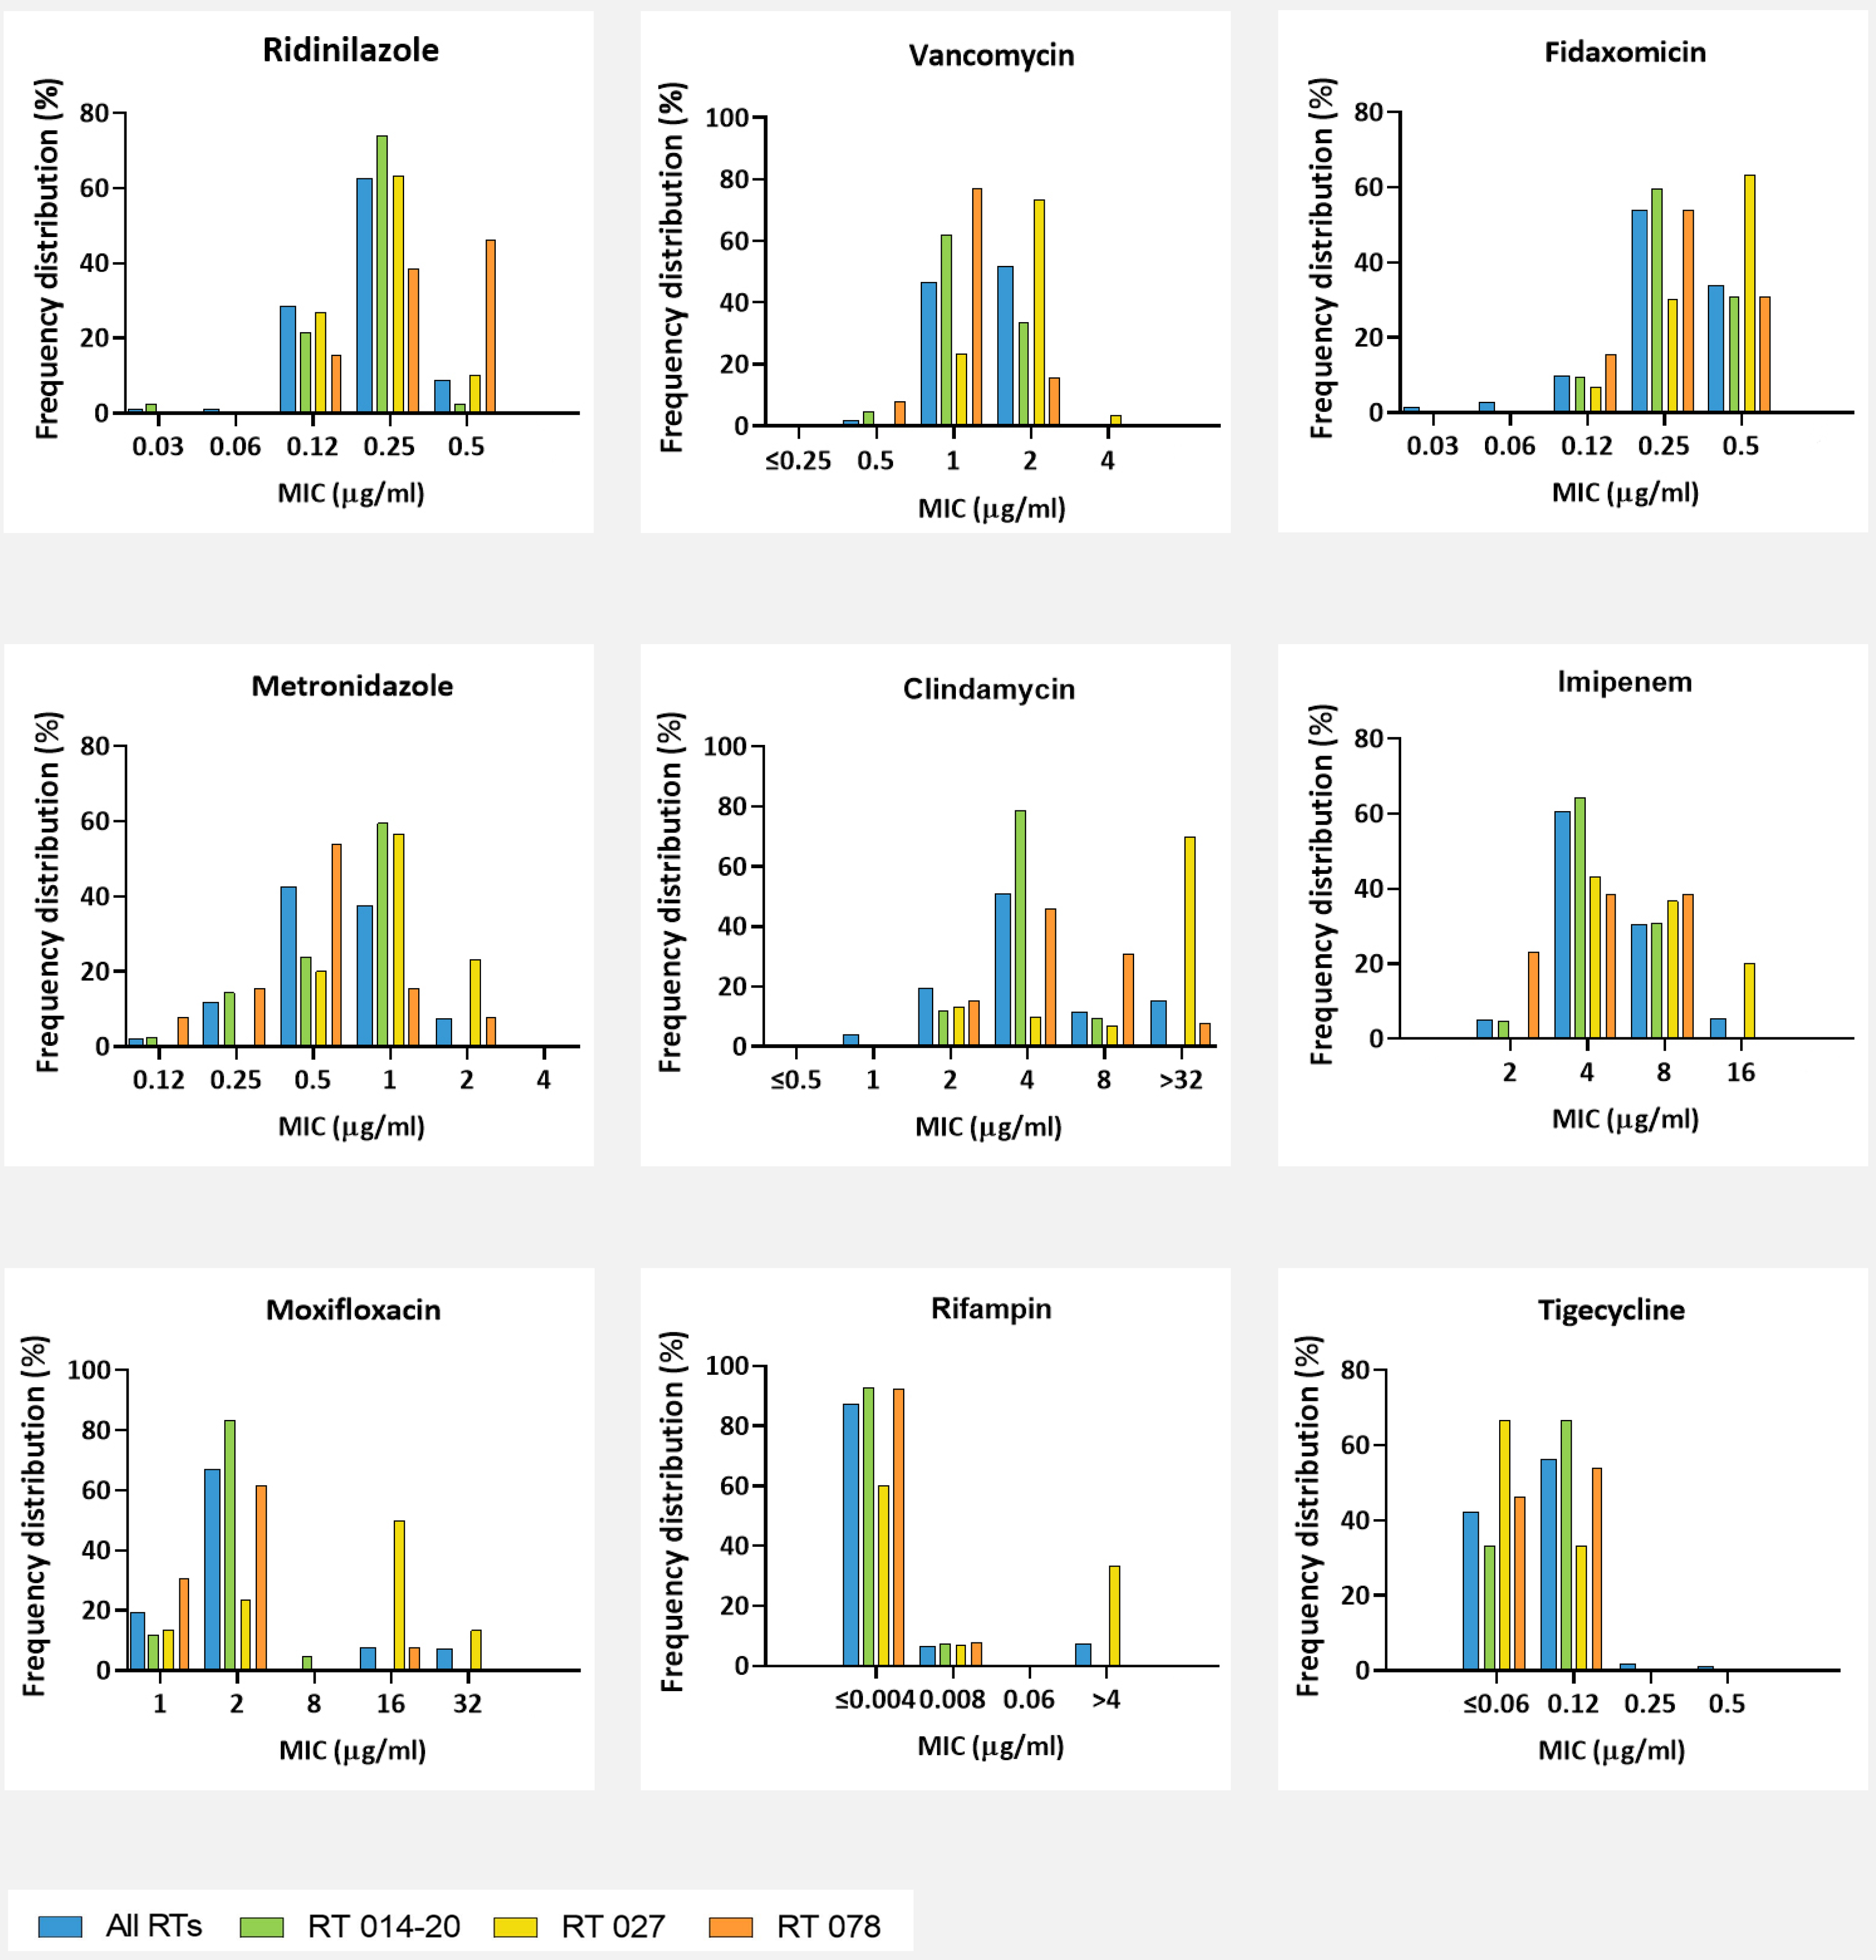

Ridinilazole
Vancomycin
Fidaxomicin
Metronidazole
Clindamycin
Imipenem
Moxifloxacin
Rifampin
Tigecycline
Frequency distribution (%)
MIC (µg/ml)
All RTs
RT 014-20
RT 027
RT 078

Ridinilazole had activity against C. *difficile* isolates with resistance to clindamycin, rifampin, moxifloxacin, imipenem, tigecycline and vancomycin based on the CLSI and EUCAST breakpoint for resistance or epidemiological values, as applicable.

Table 3. Activity of Ridinilazole and Comparators Against Isolates Demonstrating Resistance to other drugs

Isolates with Resistance to:		Ridinilazole	Vancomycin	Fidaxomicin	Metronidazole
Clindamycin N=78	MIC_{50}	**0.25**	2	0.25	1
	MIC_{90}	**0.5**	2	0.5	2
	% R CLSI	-	2.6	-	0.0%
	%R EUCAST	-	2.6%	-	1.3%
Rifampin N=45	MIC_{50}	**0.25**	2	0.5	1
	MIC_{90}	**0.25**	2	0.5	2
	% R CLSI	-	2.2%	-	0.0%
	%R EUCAST	-	2.2%	-	2.2%
Moxifloxacin N=44	MIC_{50}	**0.25**	2	0.25	1
	MIC_{90}	**0.25**	2	0.5	2
	% R CLSI	-	4.5%	-	0.0%
	%R EUCAST	-	2.6%	-	1.3%
Imipenem N=15	MIC_{50}	**0.25**	2	0.25	1
	MIC_{90}	**0.25**	2	0.5	2
	% R CLSI	-	6.7%	-	0.0%
	%R EUCAST	-	6.7%	-	0.0%
Tigecycline N=2	MICs	**0.12, 0.25**	both 2	0.06, 0.25	0.5, 2
	% R CLSI[1]	-	0.0%	-	0.0%
	%R EUCAST	-	0.0%	-	0.0%
Vancomycin N=2	MICs	**0.12, 0.25**	both 4	both 0.12	0.5, 1
	% R CLSI	-	100%	-	0.0%
	%R EUCAST	-	100.0%	-	0.0%

MIC_{50} and MIC_{90} are expressed in µg/ml; -: no CLSI or EUCAST recommended breakpoint for resistance or epidemiological cut-off values available; % R: percentage of resistant isolates based on the clinical breakpoints or based on the epidemiological cut-off value in the absence of a clinical breakpoint recommended by CLSI or EUCAST, as applicable. [1] For tigecycline the breakpoint for resistance recommended for anaerobes by the FDA was used.

CONCLUSIONS

- **Ridinilazole showed excellent in vitro activity against contemporary *C. difficile* isolates collected in the US (2020-2021) with a MIC_{90} of 0.25 µg/ml.**
- **Ridinilazole retained activity against all ribotypes including hypervirulent ribotypes 027 and 078.**
- **Ridinilazole retained activity against *C. difficile* isolates which were resistant to other antibiotics.**
- **There has been a change in ribotype distribution compared to 2016 accompanied by a reduction in *C. difficile* resistance to imipenem, and moxifloxacin.[7]**

REFERENCES

1. CDC. Antibiotic Resistance Threats in the United States, 2019. http://dx.doi.org/10.15620/cdc:82532.
2. Snydman, D. R. et al. 2015. *Antimicrob Agents Chemother* 59:6437–6443. https://doi.org/10.1128/AAC.00845-15
3. Thorpe, C. M. et al. 2019. *Antimicrob Agents Chemother* 63:e00391-19. https://doi.org/10.1128/AAC.00391-19

4. Goldstein E. J. C. et al. 2013. *Antimicrob Agents Chemother* 57:4872-4876. https://doi.org/10.1128/AAC.01136-13
5. Vickers R. J. et al. 2017. *Lancet Infect Dis.* 17:735-744; https://doi.org/10.1016/S1473-3099(17)30235-9
6. Thorpe C. M. et al. 2018. PLoS ONE 13(8):e0199810. https://doi.org/10.1371/journal.pone.0199810
7.Snydman, D. R. et al. 2020. Anaerobe 63 (2020). https://doi.org/10.1016/j.anaerobe.2020.102185

© 2022 Summit Therapeutics, Inc.;
Trademarks, registered or otherwise, are the property of their respective owners.